A First-in-Class HER2-Targeted Radiopharmaceutical Therapy:
Initial Findings From the Phase 0/1 HEAT Trial of ^{177}Lu-RAD202 in HER2+ Advanced Solid Tumors

Aviral Singh[1], Udit Nindra[2], Ian Swainson[2], **Dimitris Voliotis**[3], Veronica Wong[4]

[1]Genesis Care Murdoch, Murdoch, Australia; [2]Illawarra & Shoalhaven Cancer Care Centers, Wollongong, Australia; [3]Radiopharm Theranostics, Madison, NJ, USA; [4]Nepean Hospital, Penrith, Australia

RADIOPHARM THERANOSTICS

Abstract CT046 | Poster 6

Introduction

TARGETING HER2 RESISTANCE WITH ^{177}Lu-RAD202

- HER2-targeted therapies have transformed outcomes for HER2-expressing solid tumors, improving survival in solid tumors, including breast cancer. Nonetheless, primary and acquired resistance remain major challenges[1]

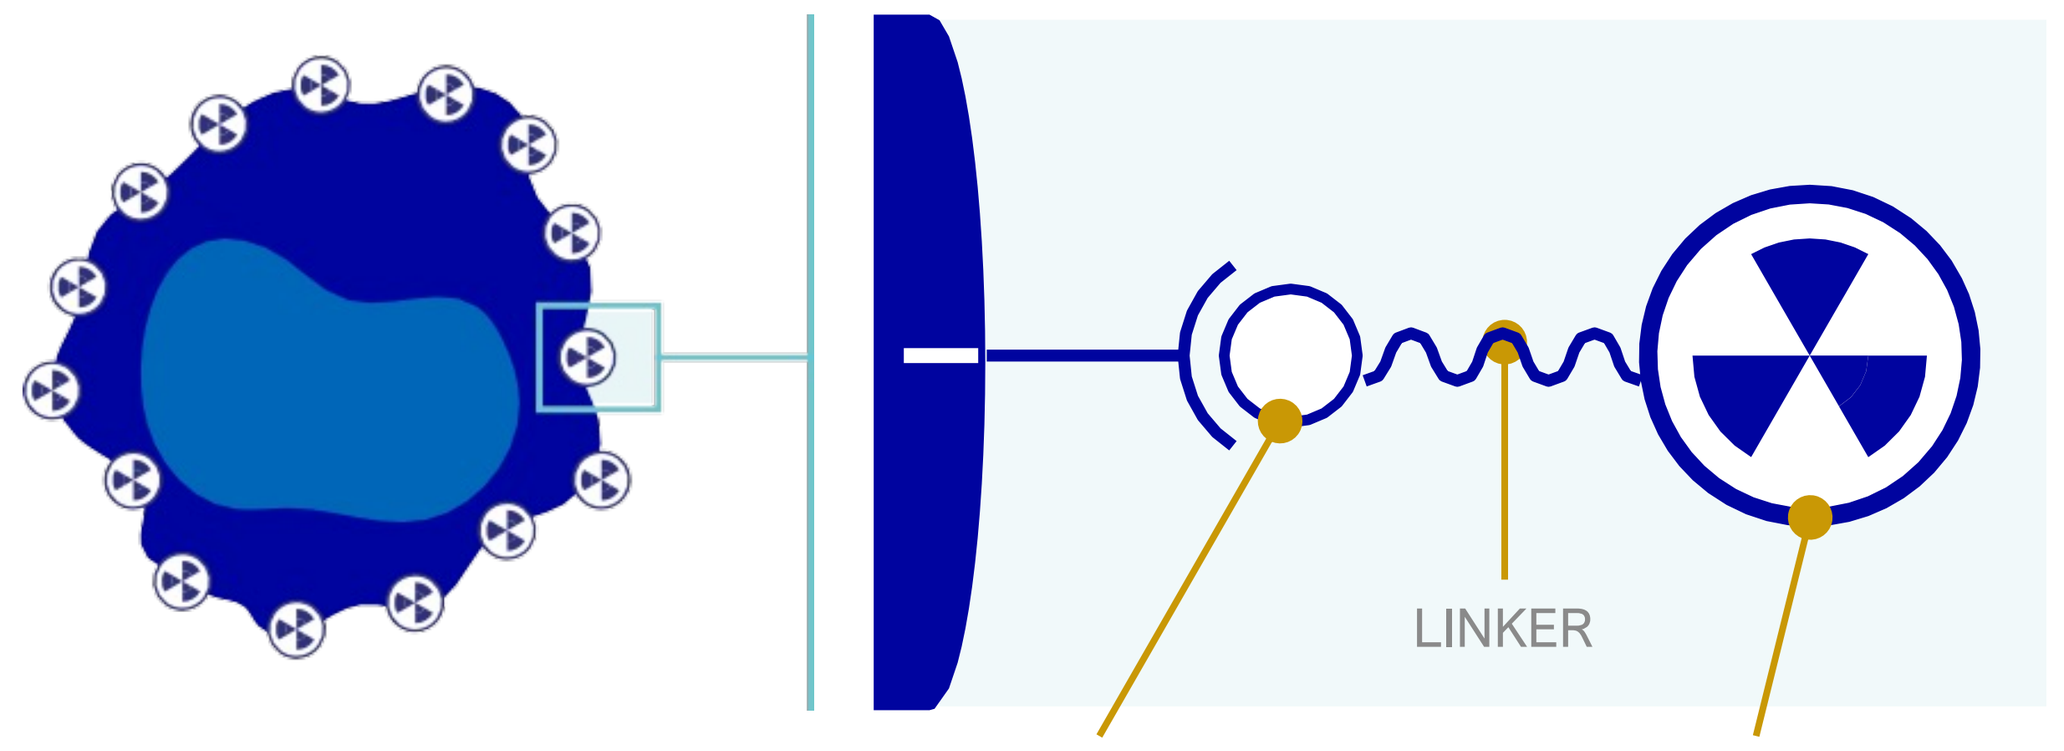
LINKER
CANCER CELL TARGET
HER2
TARGETING MOLECULE
sdAb
RADIOACTIVE ISOTOPE
177Lu

- ^{177}Lu-RAD202 is a Lutetium-177–labeled sdAb targeting HER2, developed for treating HER2-positive (HER2+) solid tumors[2]
- The small size of an sdAb enables deep tumor penetration and rapid clearance[3], while ^{177}Lu emits cytotoxic β-radiation producing a bystander effect independent of receptor density[4]
- A Phase I diagnostic study with 99mTc-RAD202 demonstrated safety, as well as favorable biodistribution and imaging characteristics (tumor targeting) in 10 patients with HER2+ breast cancer[5,6]

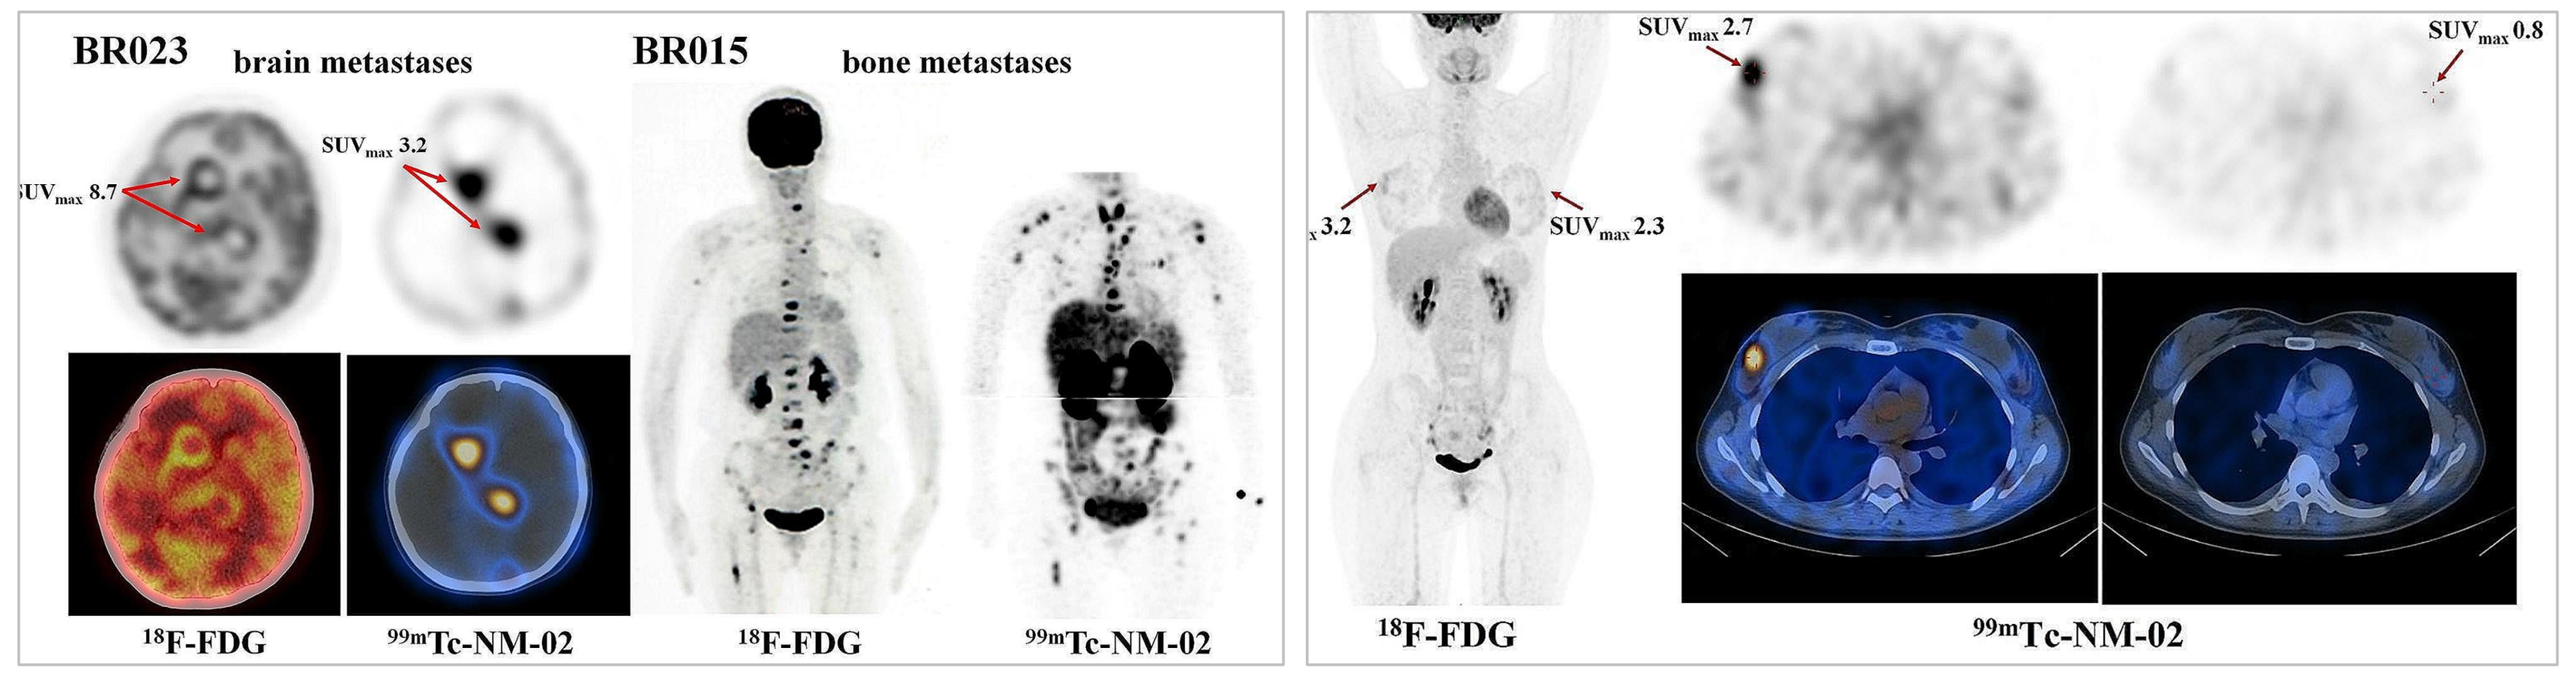
BR023 brain metastases
SUVmax 8.7
SUVmax 3.2
BR015 bone metastases
18F-FDG
99mTc-NM-02
18F-FDG
99mTc-NM-02
SUVmax 2.7
SUVmax 0.8
3.2
SUVmax 2.3
18F-FDG
99mTc-NM-02

- ^{177}Lu-RAD202 may overcome resistance to monoclonal antibody, TKI, and ADC therapies, supporting further clinical evaluation

Methods

HEAT TRIAL | Phase 0/1 Multiple Dosing Study

The HEAT Trial (HER2-Antibody Therapy With Lutetium-177; RAD202; NCT06824155) is a first-in-human, open-label, multicenter Phase 0/1 multiple dosing study of ^{177}Lu-RAD202 in HER2+ advanced solid tumors

Primary Objectives (Phase 1, Treatment):
- Safety and tolerability of ^{177}Lu-RAD202
- Recommended Phase 2 dose of ^{177}Lu-RAD202

Population:
HER2+ (IHC, ISH) a/m solid tumors

Phase 0 Imaging:
Biodistribution, PK, and radiation dosimetry of ^{177}Lu-RAD202$_{im}$ in organs of interest and tumor lesions

Phase 1 Therapeutic:
^{177}Lu-RAD202$_{tr}$ dose escalation

	Dose Level	Dose
Phase 0 (Imaging Period With ^{177}Lu-RAD202$_{im}$)	**Imaging dose**	10 mCi
Phase 1 (Treatment Period With ^{177}Lu-RAD202$_{tr}$)	**Therapeutic DL1**	30 mCi (1.1 GBq)
	DL2	75 mCi (2.7 GBq)
	DL3+	TBD

Primary objectives are to evaluate safety, clinical activity, tumor targeting and determine the recommended therapeutic dose(s)

Cohort 1 Results

PARTICIPANT SUMMARY

Participant Demographics

Participant	Age	Sex	Race	Primary Diagnosis	Number of Prior Metastatic Treatments
001	62	F	Asian	Metastatic breast cancer (infiltrating ductal carcinoma) HER2+/ER+/PR+	17
002	57	F	White	Metastatic breast cancer (large cell carcinoma) HER2+/PR+/ER-	6
003	55	F	White	Metastatic urothelial cancer (invasive papillary carcinoma) HER2+	4

SAFETY DATA

Treatment-Emergent Adverse Events	Patients, n	CTCAE Grade
Pain	2	Grade 1
ALT increased	1	
AST increased	1	
Cough	1	
Dry mouth	1	
Dysgeusia	1	
Fatigue	1	
Flu-like symptoms	1	
Gastroesophageal reflux disease	1	
Hot flashes	1	
Insomnia	1	
Lymph leakage	1	
Oozing from breast	1	
Pleural effusion	1	
Skin tear	1	
Worsening of anxiety	1	Grade 2
AST increased	1	
Pain lower back	1	
Fracture	1	
Pruritus	1	
Dyspnea	1	Grade 3

^{177}Lu-RAD202 was generally well tolerated, with predominantly Grade 1-2 TEAEs.
No dose-limiting toxicities or treatment discontinuations due to AEs were observed.
Data represents the number of patients that experienced a particular TEAE.
Total patient population N = 3.

Serious Adverse Events	Patients, n	CTCAE Grade
Dyspnea	1	Grade 3
Pharyngitis	1	
Neutrophil count decreased	1	Grade 4

All events were unrelated to study drug as deemed by investigator judgement.

TUMOR RESPONSE

Swimmer Plot of Response Over Time

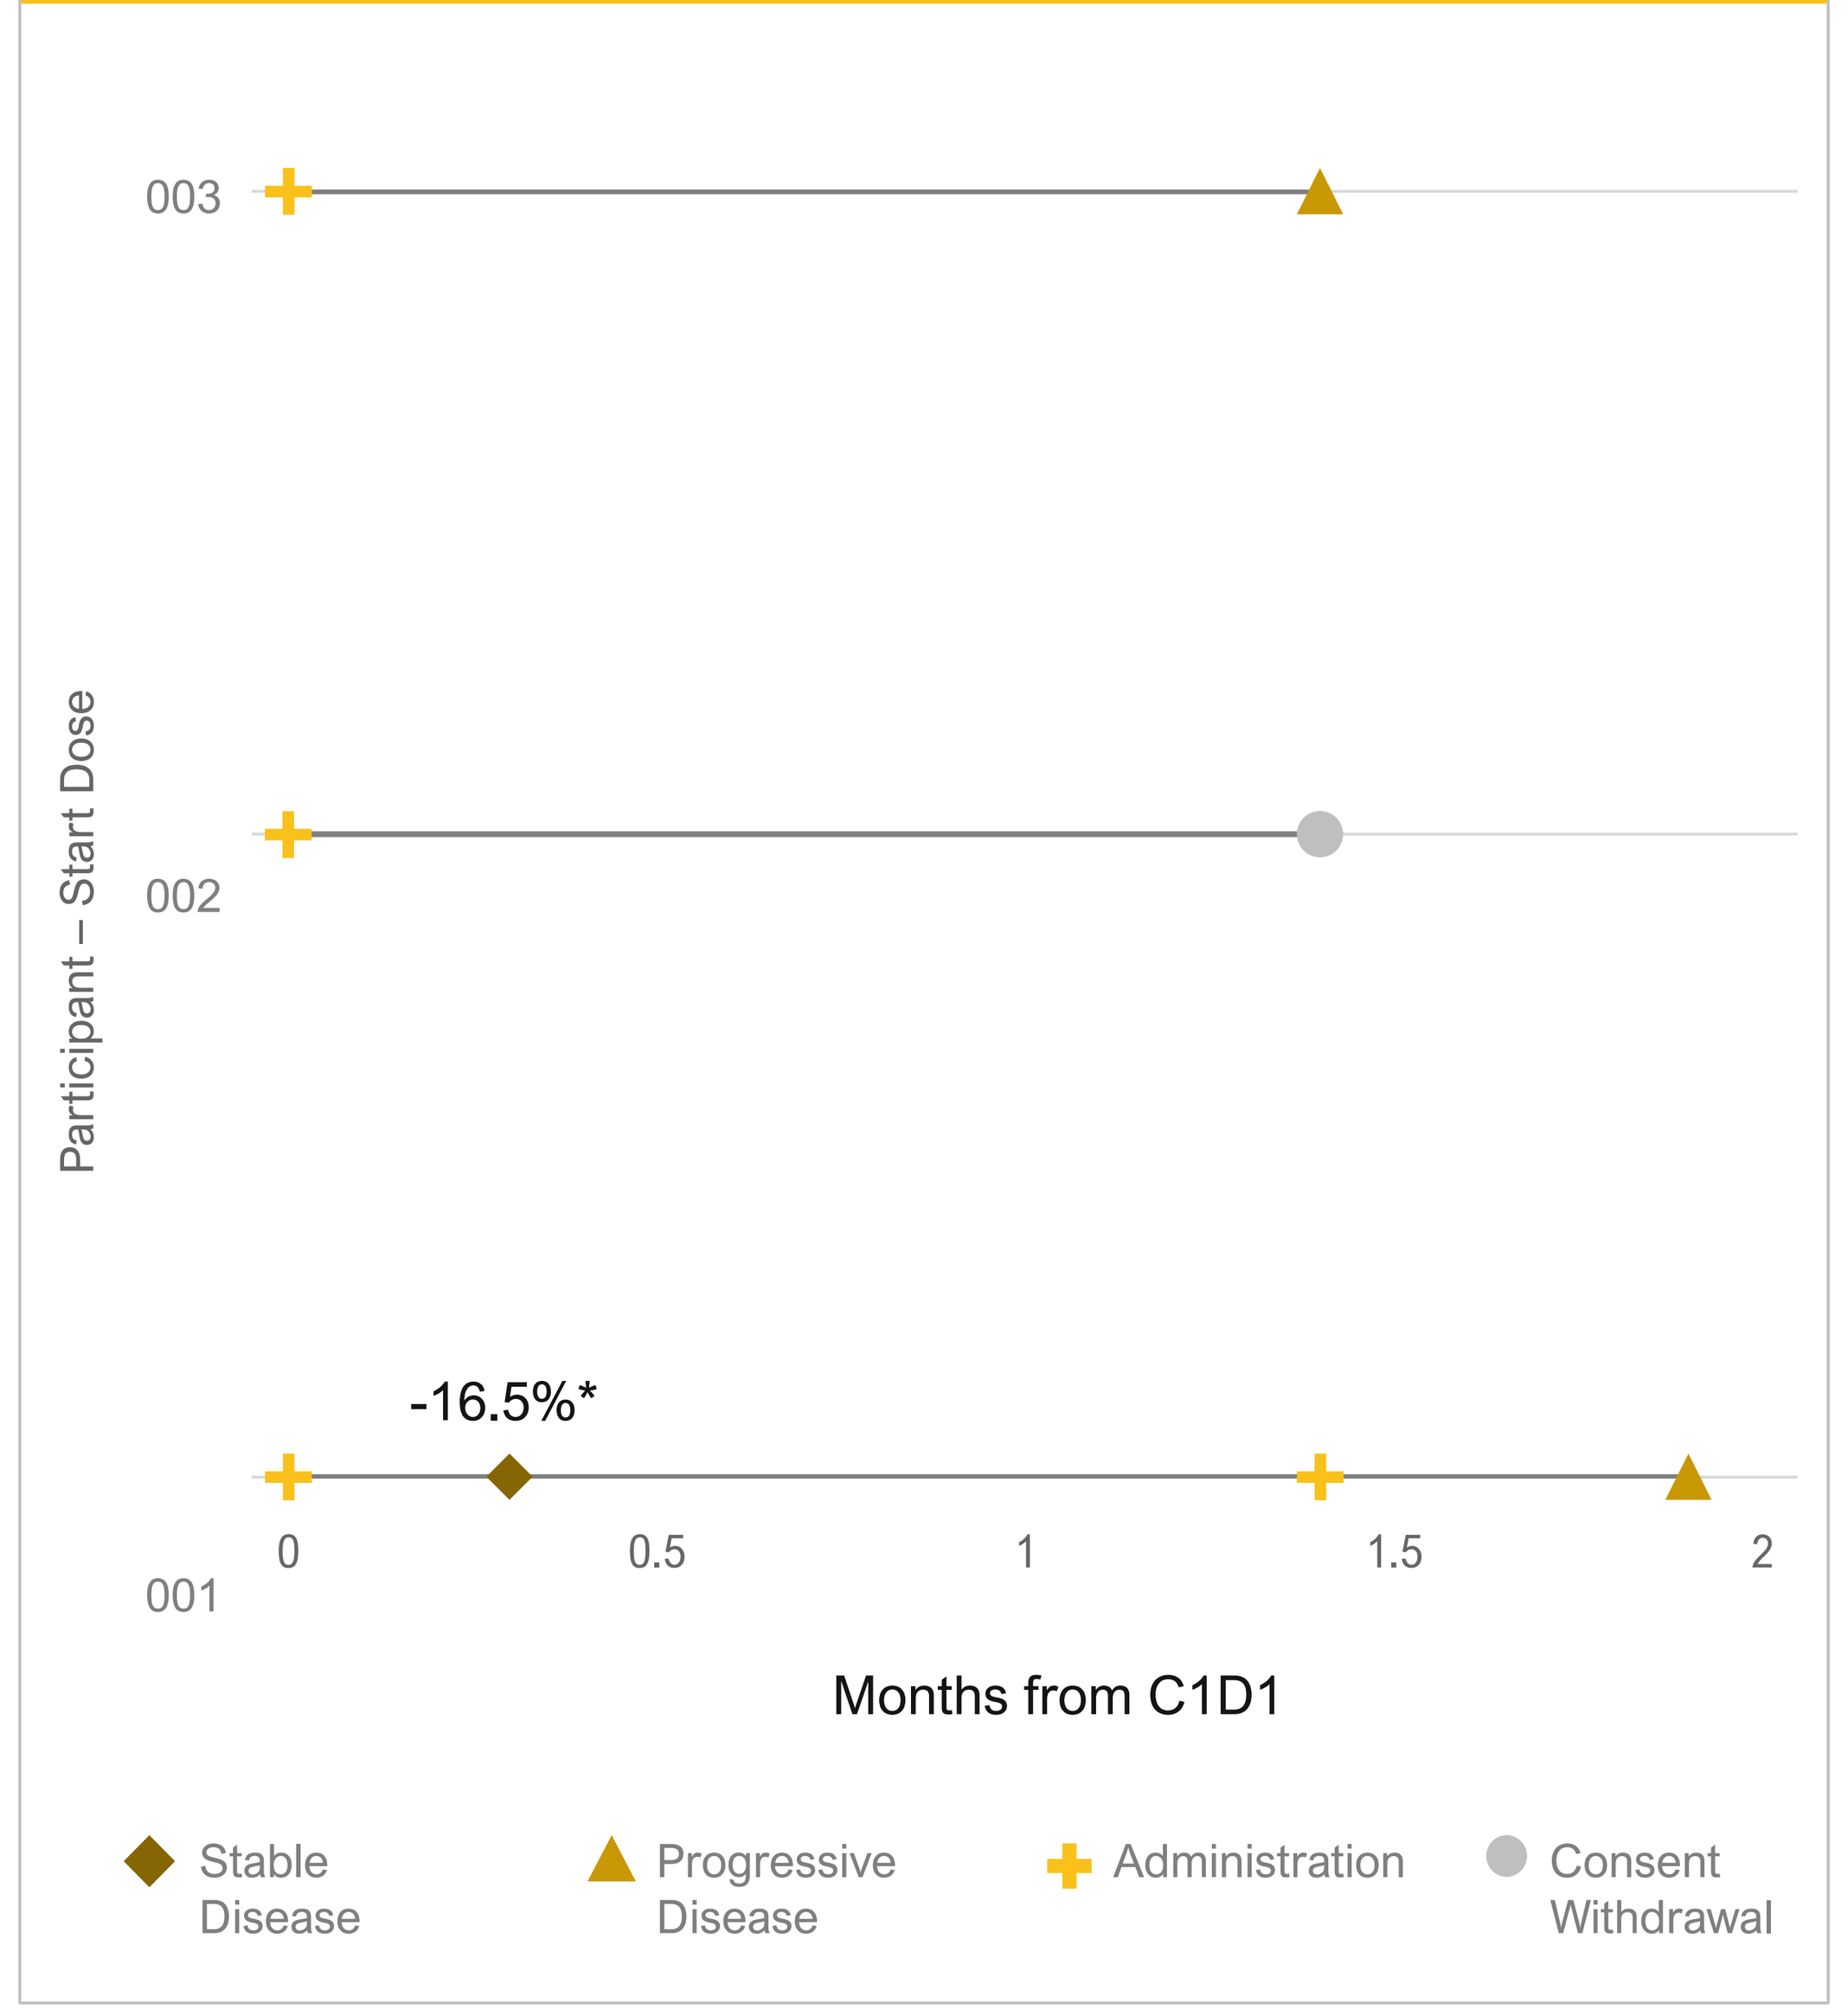
003
002
001
Participant – Start Dose
-16.5%*
0
0.5
1
1.5
2
Months from C1D1
Stable Disease
Progressive Disease
Administration
Consent Withdrawal

*Best overall response was stable disease, with a reduction in target lesion size of -16.5%. For the other two heavily pre-treated patients, treatment duration was limited due to early discontinuation (progression or withdrawal), therefore response could not be fully assessed. These early findings demonstrate initial signs of antitumor activity with ^{177}Lu-RAD202.

BIODISTRIBUTION & DOSIMETRY | Phase 1

Tumor Uptake

Participant	Cycle	Dose	Lesion	Tumor Type	Absorbed Dose (Gy) with PVC†
001	C1	30 mCi	ROI-2	Breast	**3.57**
002			ROI-11	Breast	**2.96**
003			ROI-11	Bladder	**1.23**

Significant tumor uptake at 30 mCi, especially in breast cancer participants.
†Density of lesion: soft tissue = 1.0 g/mL, bone = 1.3 g/mL.

Lesion Dosimetry

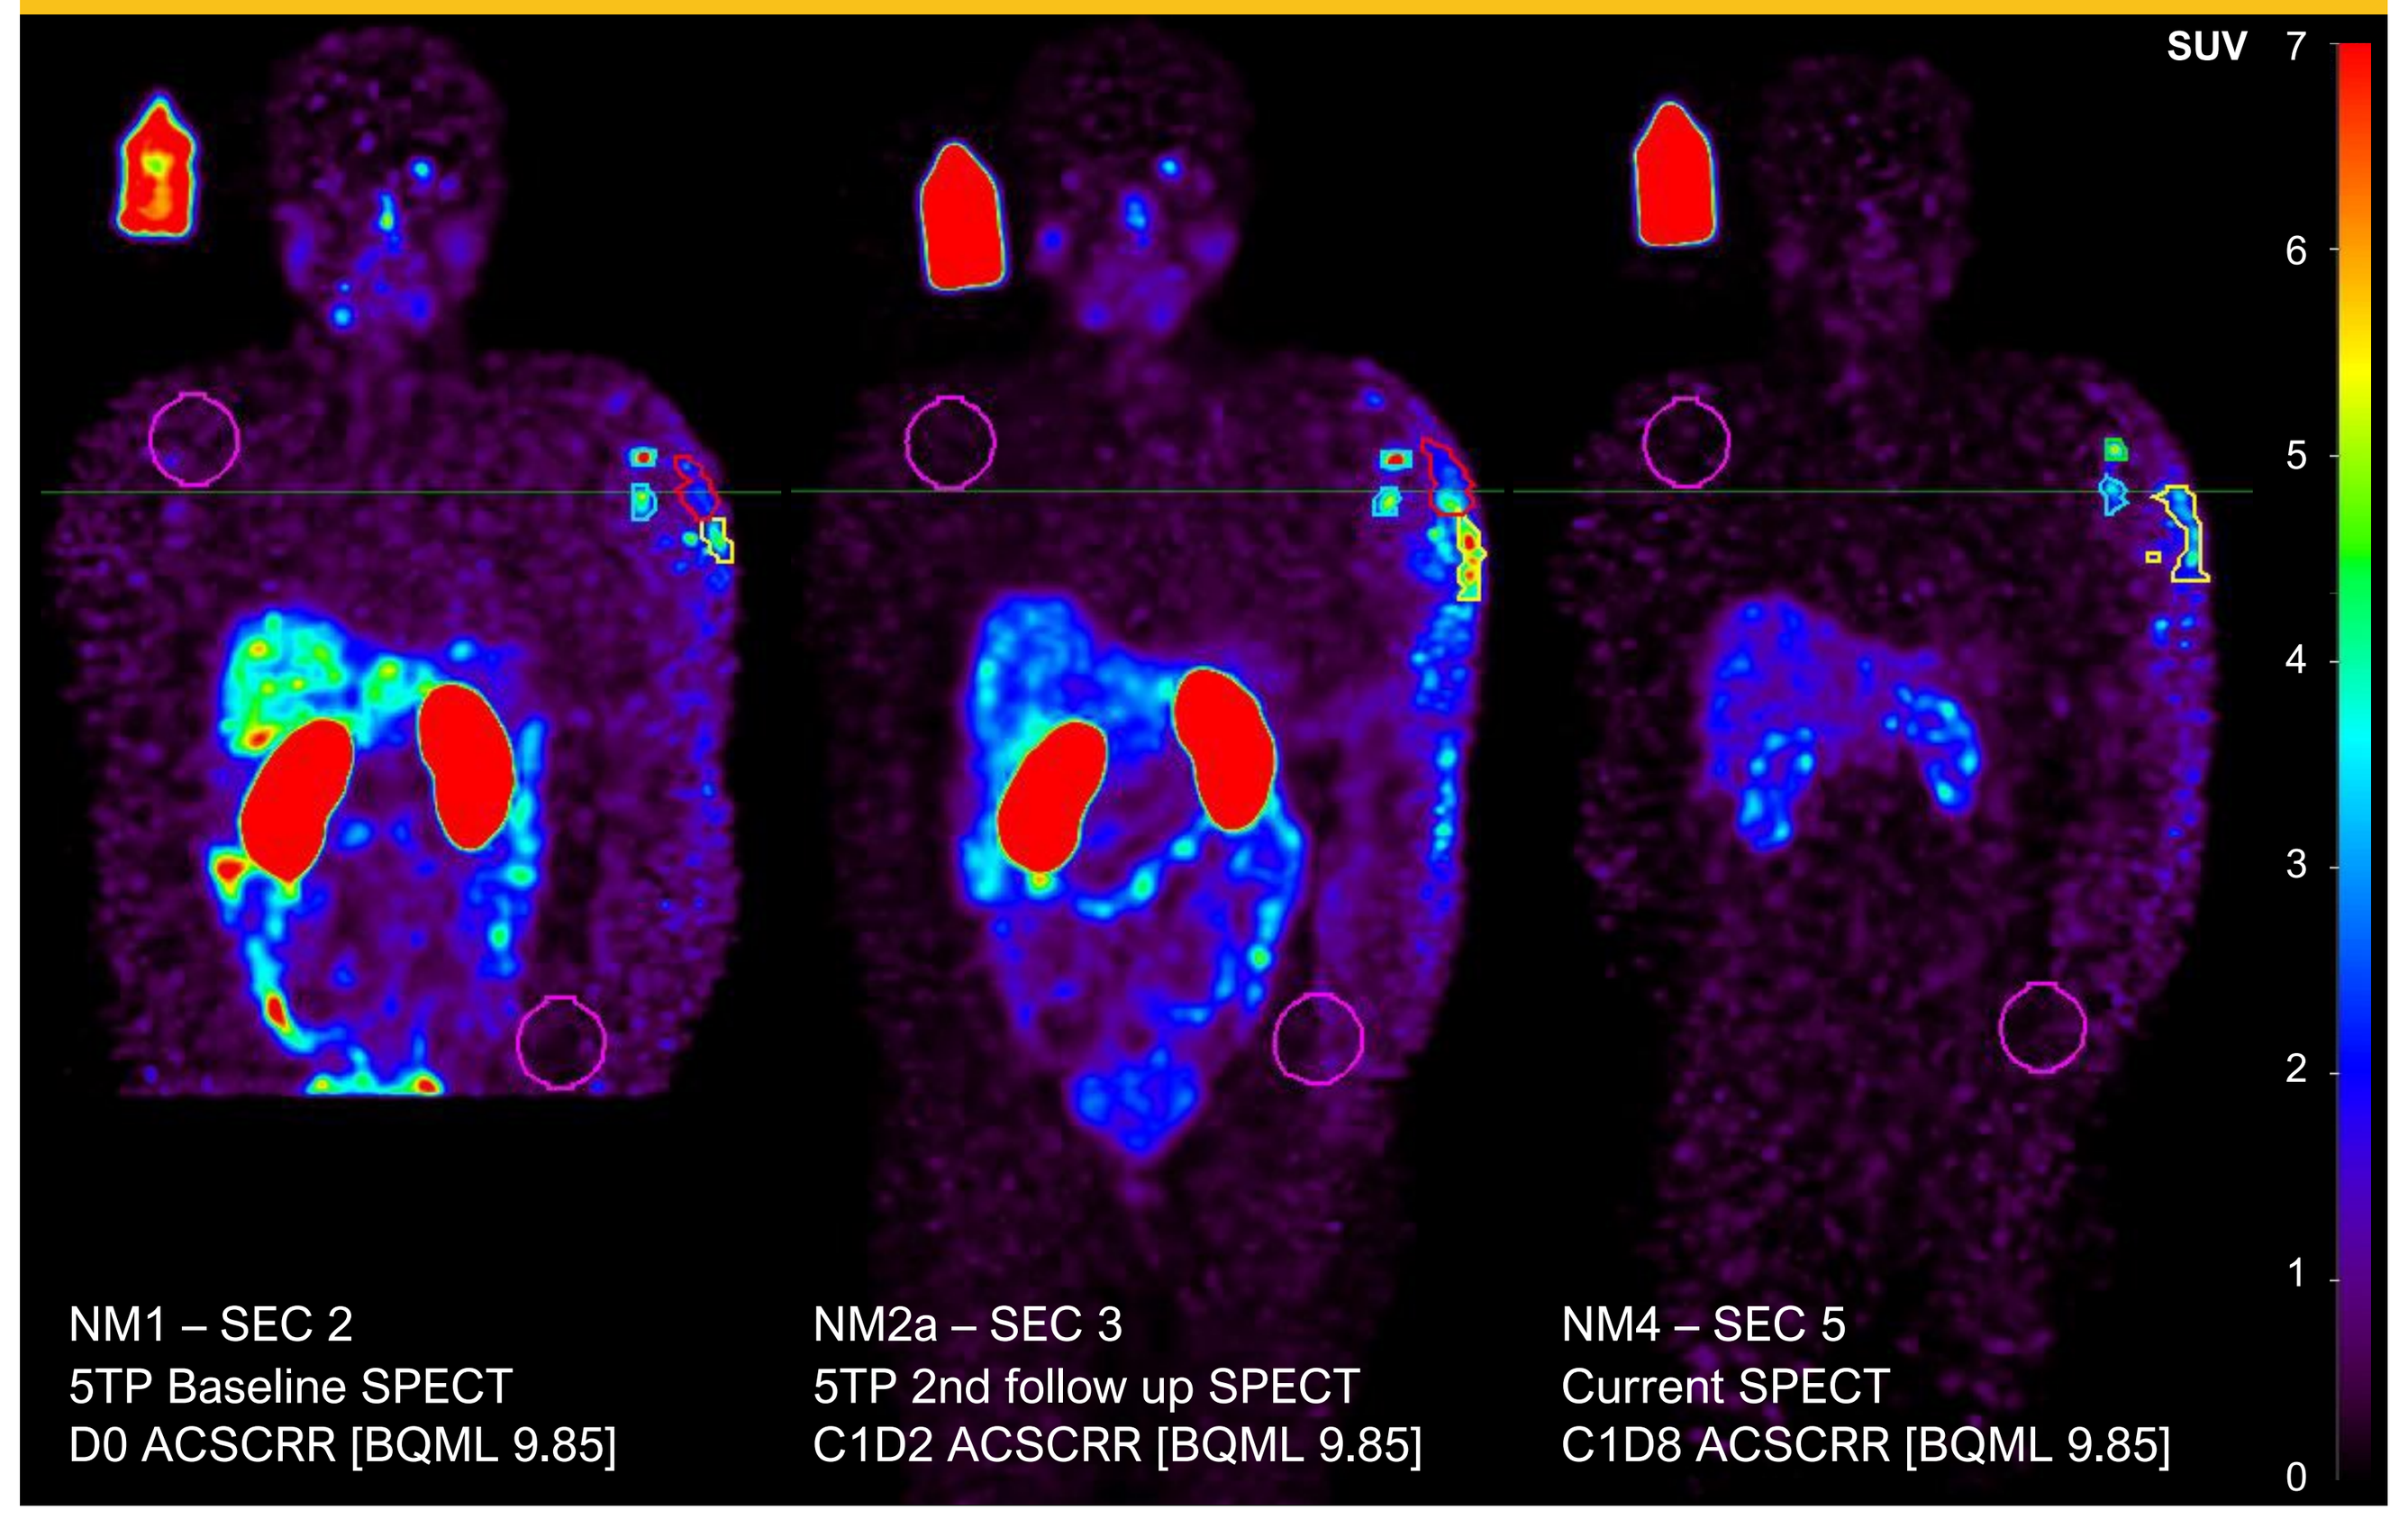
SUV
7
6
5
4
3
2
1
0
NM1 – SEC 2
5TP Baseline SPECT
D0 ACSCRR [BQML 9.85]
NM2a – SEC 3
5TP 2nd follow up SPECT
C1D2 ACSCRR [BQML 9.85]
NM4 – SEC 5
Current SPECT
C1D8 ACSCRR [BQML 9.85]

Participant 1: Selective uptake of ^{177}Lu-RAD202 in metastatic skin and subcutaneous lesions.

Organ Dosimetry

	Absorbed Dose (Gy)				
Treatment	**C1**	**C2‡**	**C1**	**C1**	**Average**
Participant	**001**	**001**	**002**	**003**	
Stomach wall	0.054	0.073	0.093	0.066	0.071
Heart wall	0.153	0.277	0.165	0.200	0.199
Kidneys	5.757	3.098	2.435	2.171	3.365
Liver	0.371	0.658	0.782	0.422	0.558
Lungs	0.114	0.166	0.143	0.120	0.136
Salivary glands	0.149	0.303	0.087	0.132	0.168
Red marrow	0.100	0.115	0.114	0.205	0.133
Spleen	0.197	0.257	0.259	0.233	0.236
Thyroid	0.088	0.155	0.088	0.133	0.116

Organ-level dosimetry demonstrated a favorable safety profile and was within expected and clinically acceptable ranges, supporting continued dose escalation.
‡C2 dosimetry, based on the single-time-point dosimetry approach, relies on C1's mono-exponential clearance and the same organ effective half-life, resulting in increased uncertainty in C2 estimates.

Conclusion

These early first-in-human data demonstrate high tumor uptake and favorable safety of ^{177}Lu-RAD202, supporting continued dose escalation. This HER2-targeted radiotherapeutic potentially offers a novel treatment approach for HER2+ solid tumors, including breast, non–small cell lung, and gastric cancer. Results from DL2 (75 mCi) and higher will be presented at a future congress.

Keywords: radioimmunotherapy (mandatory), HER2-positive breast cancer, treatment-resistant HER2-positive cancer, radiopharmaceutical.

Abbreviations: ADC, antibody-drug conjugates; ALT, alanine aminotransferase; AST, aspartate aminotransferase; a/m, advanced/metastatic; CX, cycle X; CTCAE, Common Terminology Criteria for Adverse Events; DL, dose level; DX, day X; ER, estrogen receptor; Gy, gray; HER2, human epidermal growth factor receptor 2; IHC, immunohistochemistry; im, intramuscular; ISH, in situ hybridization; PK, pharmacokinetic; PR, progesterone receptor; PVC, partial volume correction; ROI, region of interest; SEC, section; sdAb, camelid single-domain antibody; SPECT, single-photon emission computed tomography TBD, to be determined; TEAE, treatment-emergent adverse event; TKI, tyrosine kinase inhibitor; tr, treatment dose.

References: 1. Schlam et al, *Cancers* (*Basel*) (2022). 2. Altunay et al, *Sci Rep* (2025). 3. Poty et al, *J Nucl Med* (2024). 4. Ferro-Flores et al, *Int J Mol Sci* (2025). 5. Zhao et al, *Br Canc Res* (2024). 6. Zhao et al, *Mol Pharmaceut* (2021).

Presented by Dimitris Voliotis at the American Association for Cancer Research (AACR) | April 17-22, 2026 in San Diego, CA

Disclosures: D.V. is an employee of Radiopharm Theranostics. A.S., U.N., I.S., and V.W. have nothing to disclose.